A Phase 1, Single-center, Open-label, Dose-rising Clinical Trial to Evaluate the Pharmacokinetics, Safety and Tolerability of Treprostinil Inhalation Powder (TreT) in Healthy Normal Volunteers

Peter M Smith, PharmD[1]; Catherine B Watkins, PharmD[1]; Marshall Grant, PhD[2]; Kelly Kraft, PhD[2]
[1]United Therapeutics, Research Triangle Park, NC, USA; [2]MannKind Corporation, Danbury, CT, USA

INTRODUCTION

- United Therapeutics is developing a combination drug-device product consisting of a dry powder formulation of treprostinil (TreT) and a small, portable, dry powder inhaler to treat pulmonary arterial hypertension (PAH).
- PAH, defined as an elevation in pulmonary arterial pressure and pulmonary vascular resistance, is a severe hemodynamic abnormality associated with a variety of diseases and syndromes.[1]
- This combination product, TreT, is a change in dosage form for treprostinil from an FDA-approved solution for oral inhalation (Tyvaso®), to a dry powder for oral inhalation.
- In addition to treprostinil, the dry powder contains the inhalation excipient fumaryl diketopiperazine (FDKP), which is an inactive excipient present in Afrezza, an FDA-approved drug product.
- Treprostinil is a chemically stable tricyclic analogue of PGI2. The pharmacology of treprostinil has been extensively characterized in well-established models, all confirming the suitability of the drug for the treatment of PAH by subcutaneous, IV, inhaled (as treprostinil sodium), or oral (as treprostinil diolamine) routes of administration.

Figure 1. TreT Dry Powder Inhaler

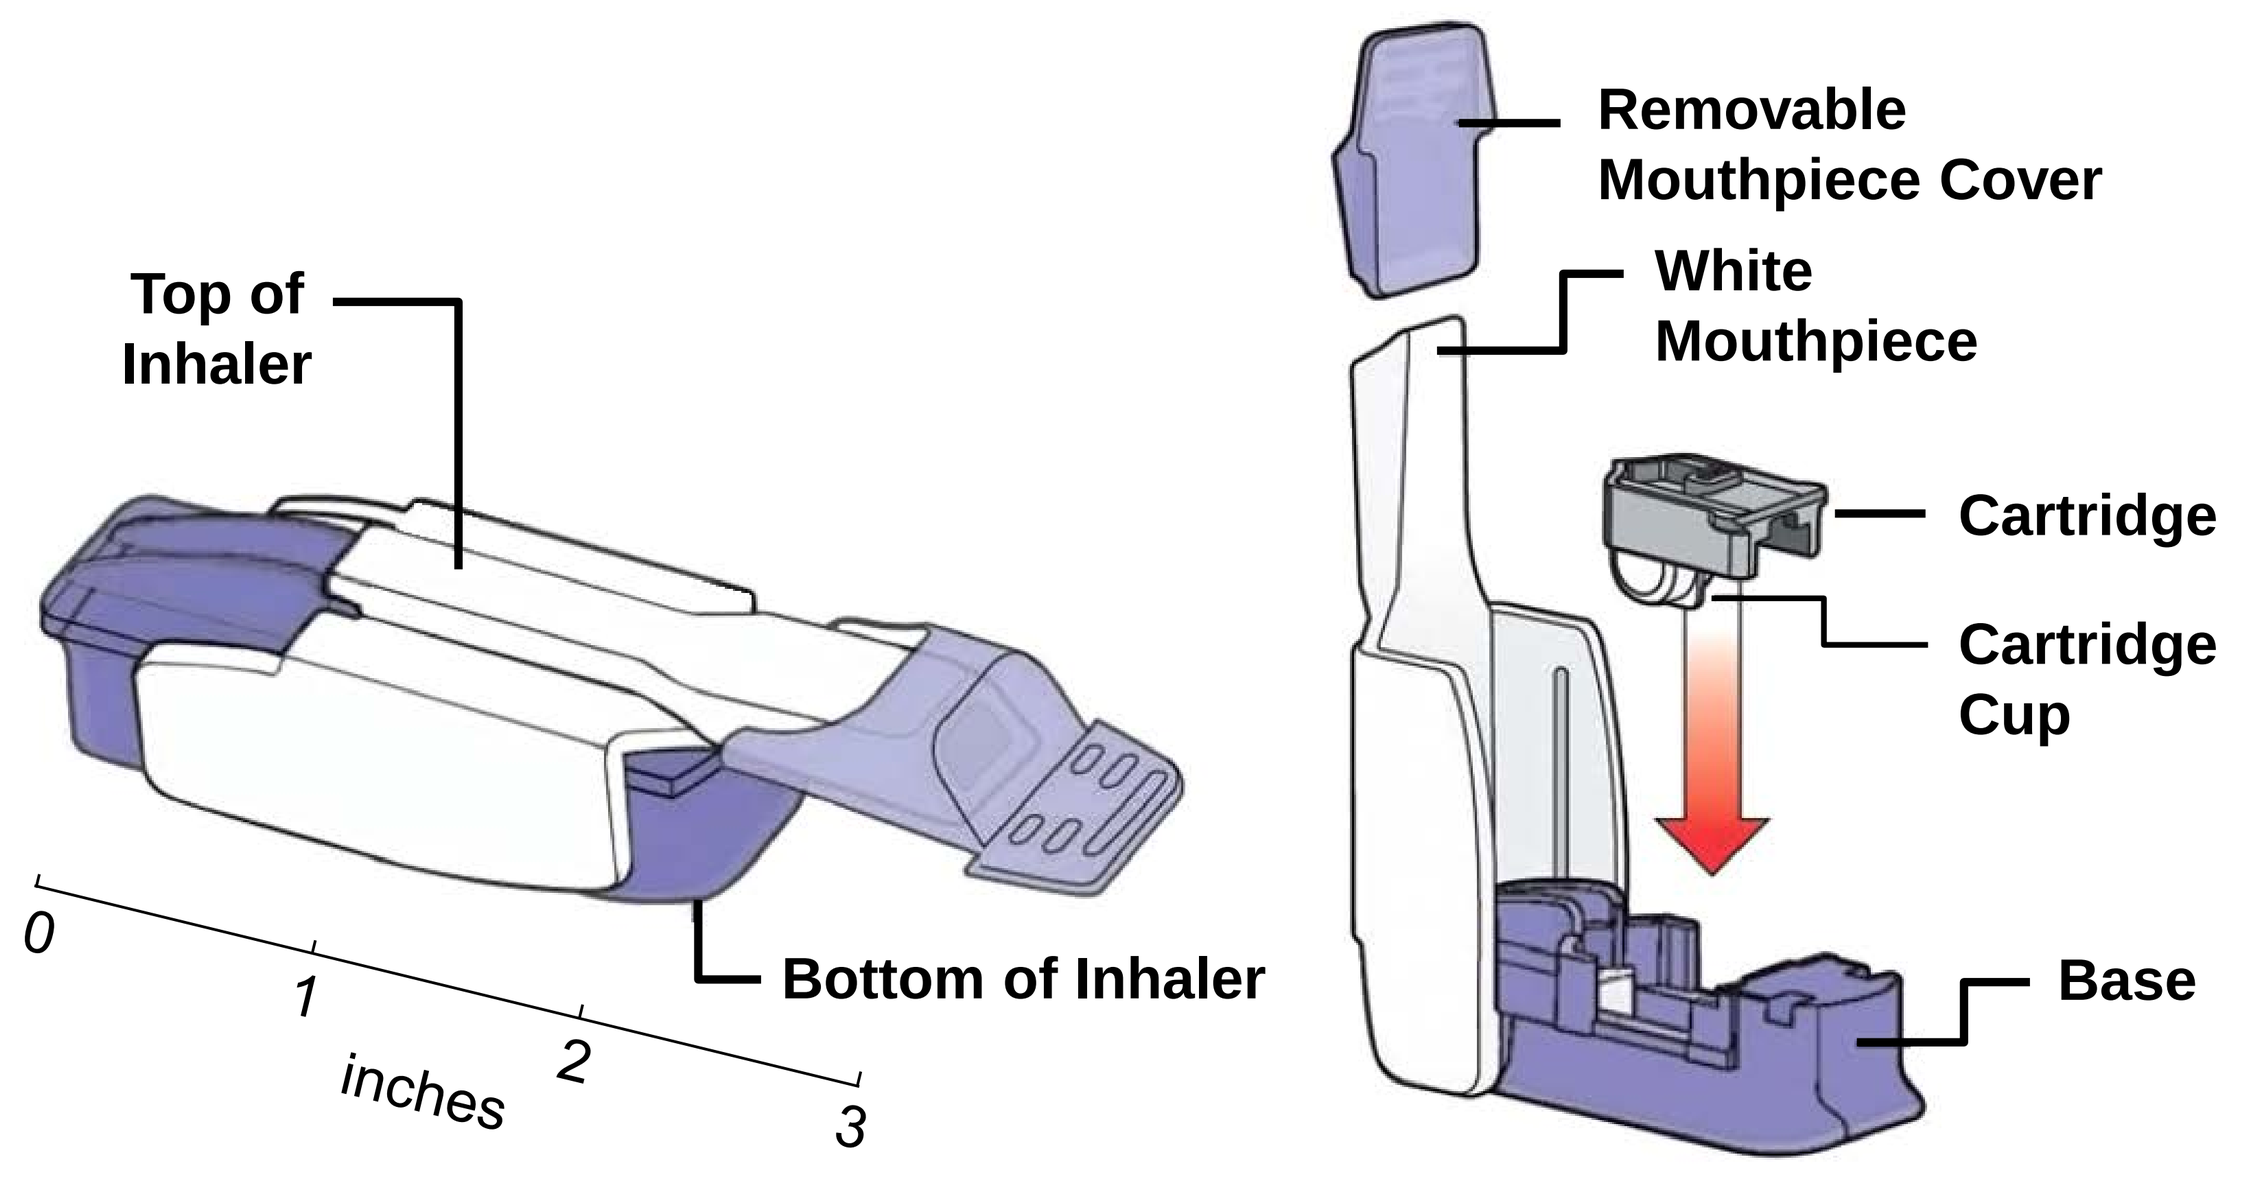
Top of Inhaler
Removable Mouthpiece Cover
White Mouthpiece
Cartridge
Cartridge Cup
Bottom of Inhaler
Base
0
1
2
3
inches

OBJECTIVES

- The primary objective was to evaluate the safety and tolerability of TreT in healthy normal volunteers (HNVs).
- The secondary objectives were to evaluate systemic exposure and pharmacokinetics (PK) of treprostinil in HNVs when delivered as TreT and to evaluate the dose proportionality of increasing doses of TreT in HNVs.

METHODS

Study Design

- This was an open-label, single ascending dose study in HNVs who were sequentially assigned to 6 cohorts of ascending dose levels of TreT (30, 60, 90, 120, 150, and 180 µg).
- Each subject received 1 dose of TreT by oral inhalation with the dry powder inhaler during the Treatment Period.
- The safety and tolerability of TreT was evaluated in each sequential cohort prior to escalating the dose for the next cohort.
- Blood samples were obtained before TreT administration and at selected times through 480 minutes post dose.
- Safety assessments included the incidence and severity of adverse events (AEs) and changes in clinical chemistry, hematology, urinalysis, vital signs, physical examinations, ECGs, and pulmonary function tests (PFTs).

Analysis

- Blood samples were analyzed for treprostinil using a validated analytical method and PK parameters were calculated using noncompartmental methods.

RESULTS

- A total of 36 HNVs were dosed and included in the pharmacokinetic and statistical analyses.
- Bioanalysis data confirmed that the treprostinil plasma concentrations and exposure for TreT achieved clinically relevant concentrations comparable to those observed in historical Tyvaso® single-dose clinical studies. 48 µg TreT corresponds to approximately 54 µg Tyvaso®, the labeled target dose.
- Treprostinil exposure with TreT increased in a linear manner with increasing dose.
- The most frequently reported AEs were cough (n=11, 30.6%) and headache (n=8, 22%).
- There were no severe AEs, serious AE (SAEs), or deaths during this study.
- No clinically significant abnormalities on oropharyngeal examinations, clinical laboratory evaluations, electrocardiograms (ECGs), or PFTs (spirometry) were observed.
- 150 µg was deemed the highest tolerated dose based on a protocol defined stopping rule met in the 180 µg cohort.

Table 1. Subject Demographics

Parameter	Overall (N=36)
Age (years)	36.0
Male	16 (44.4%)
Female	20 (55.6%)
Ethnicity	
Hispanic or Latino	24 (66.7%)
Not Hispanic or Latino	12 (33.3%)
Race	
White	26 (72.2%)
Black or African American	9 (25.0%)
American Indian or Alaska Native	1 (2.8%)
BMI (kg/m²)	26.89

RESULTS (cont'd)

Figure 2. Mean Plasma Treprostinil Concentration-time Profiles after Single-dose Administration of 30 mcg to 180 mcg TreT

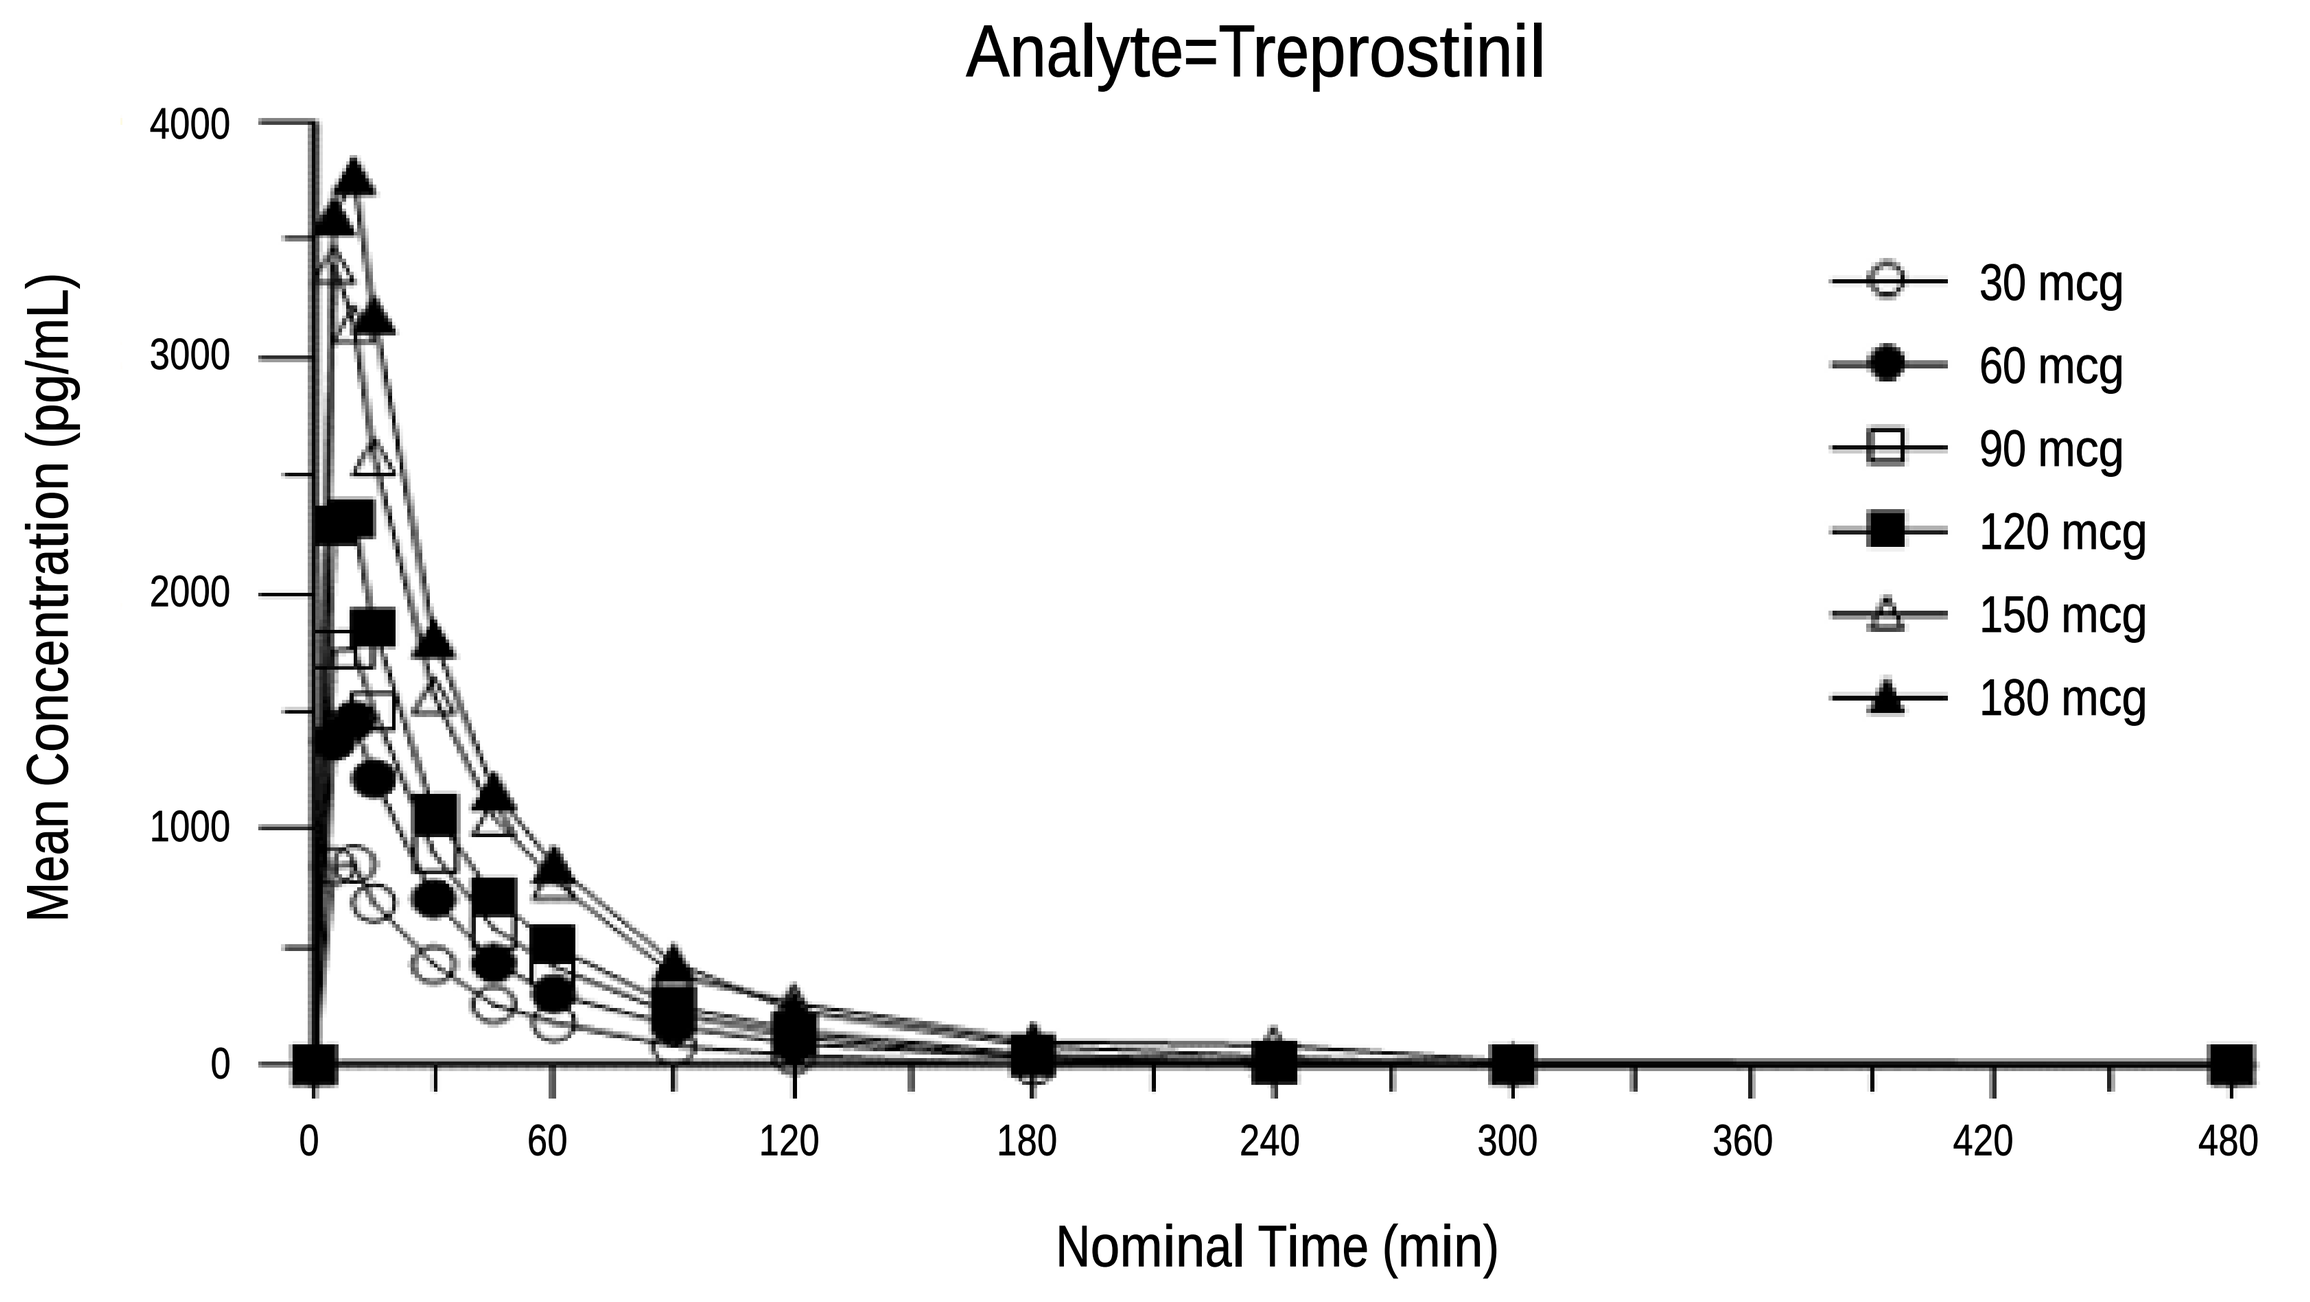
Analyte=Treprostinil
Mean Concentration (pg/mL)
0
1000
2000
3000
4000
Nominal Time (min)
0
60
120
180
240
300
360
420
480
30 mcg
60 mcg
90 mcg
120 mcg
150 mcg
180 mcg

Table 2. TreT PK Results

PK Parameter	Treprostinil (mcg)					
	30	60	90	120	150	180
AUC_{0-8} (pg-hr/mL)	578.3	1041.7	1318.3	1645.0	2650.0	2683.3
Cmax (pg/mL)	893.0	1460.0	1880.0	2360.0	3430.0	3810.0
Tmax (hr)	0.13	0.17	0.13	0.14	0.11	0.14
$T_{1/2}$ (hr)	0.45	0.57	0.54	0.60	0.84	0.69

Table 3. Summary of Adverse Events Occurring in More than 1 Subject

Adverse Event Term	Number (% of Subjects)
Cough	11 (31)
Headache	8 (22)
Nausea	7 (19)
Dizziness	7 (19)
Throat Irritation	4 (11)
Oxygen Saturation Decreased	2 (6)
Hyperhidrosis	2 (6)

RESULTS (cont'd)

Figure 3. AUC Comparison Between TreT and Tyvaso®[2,3]

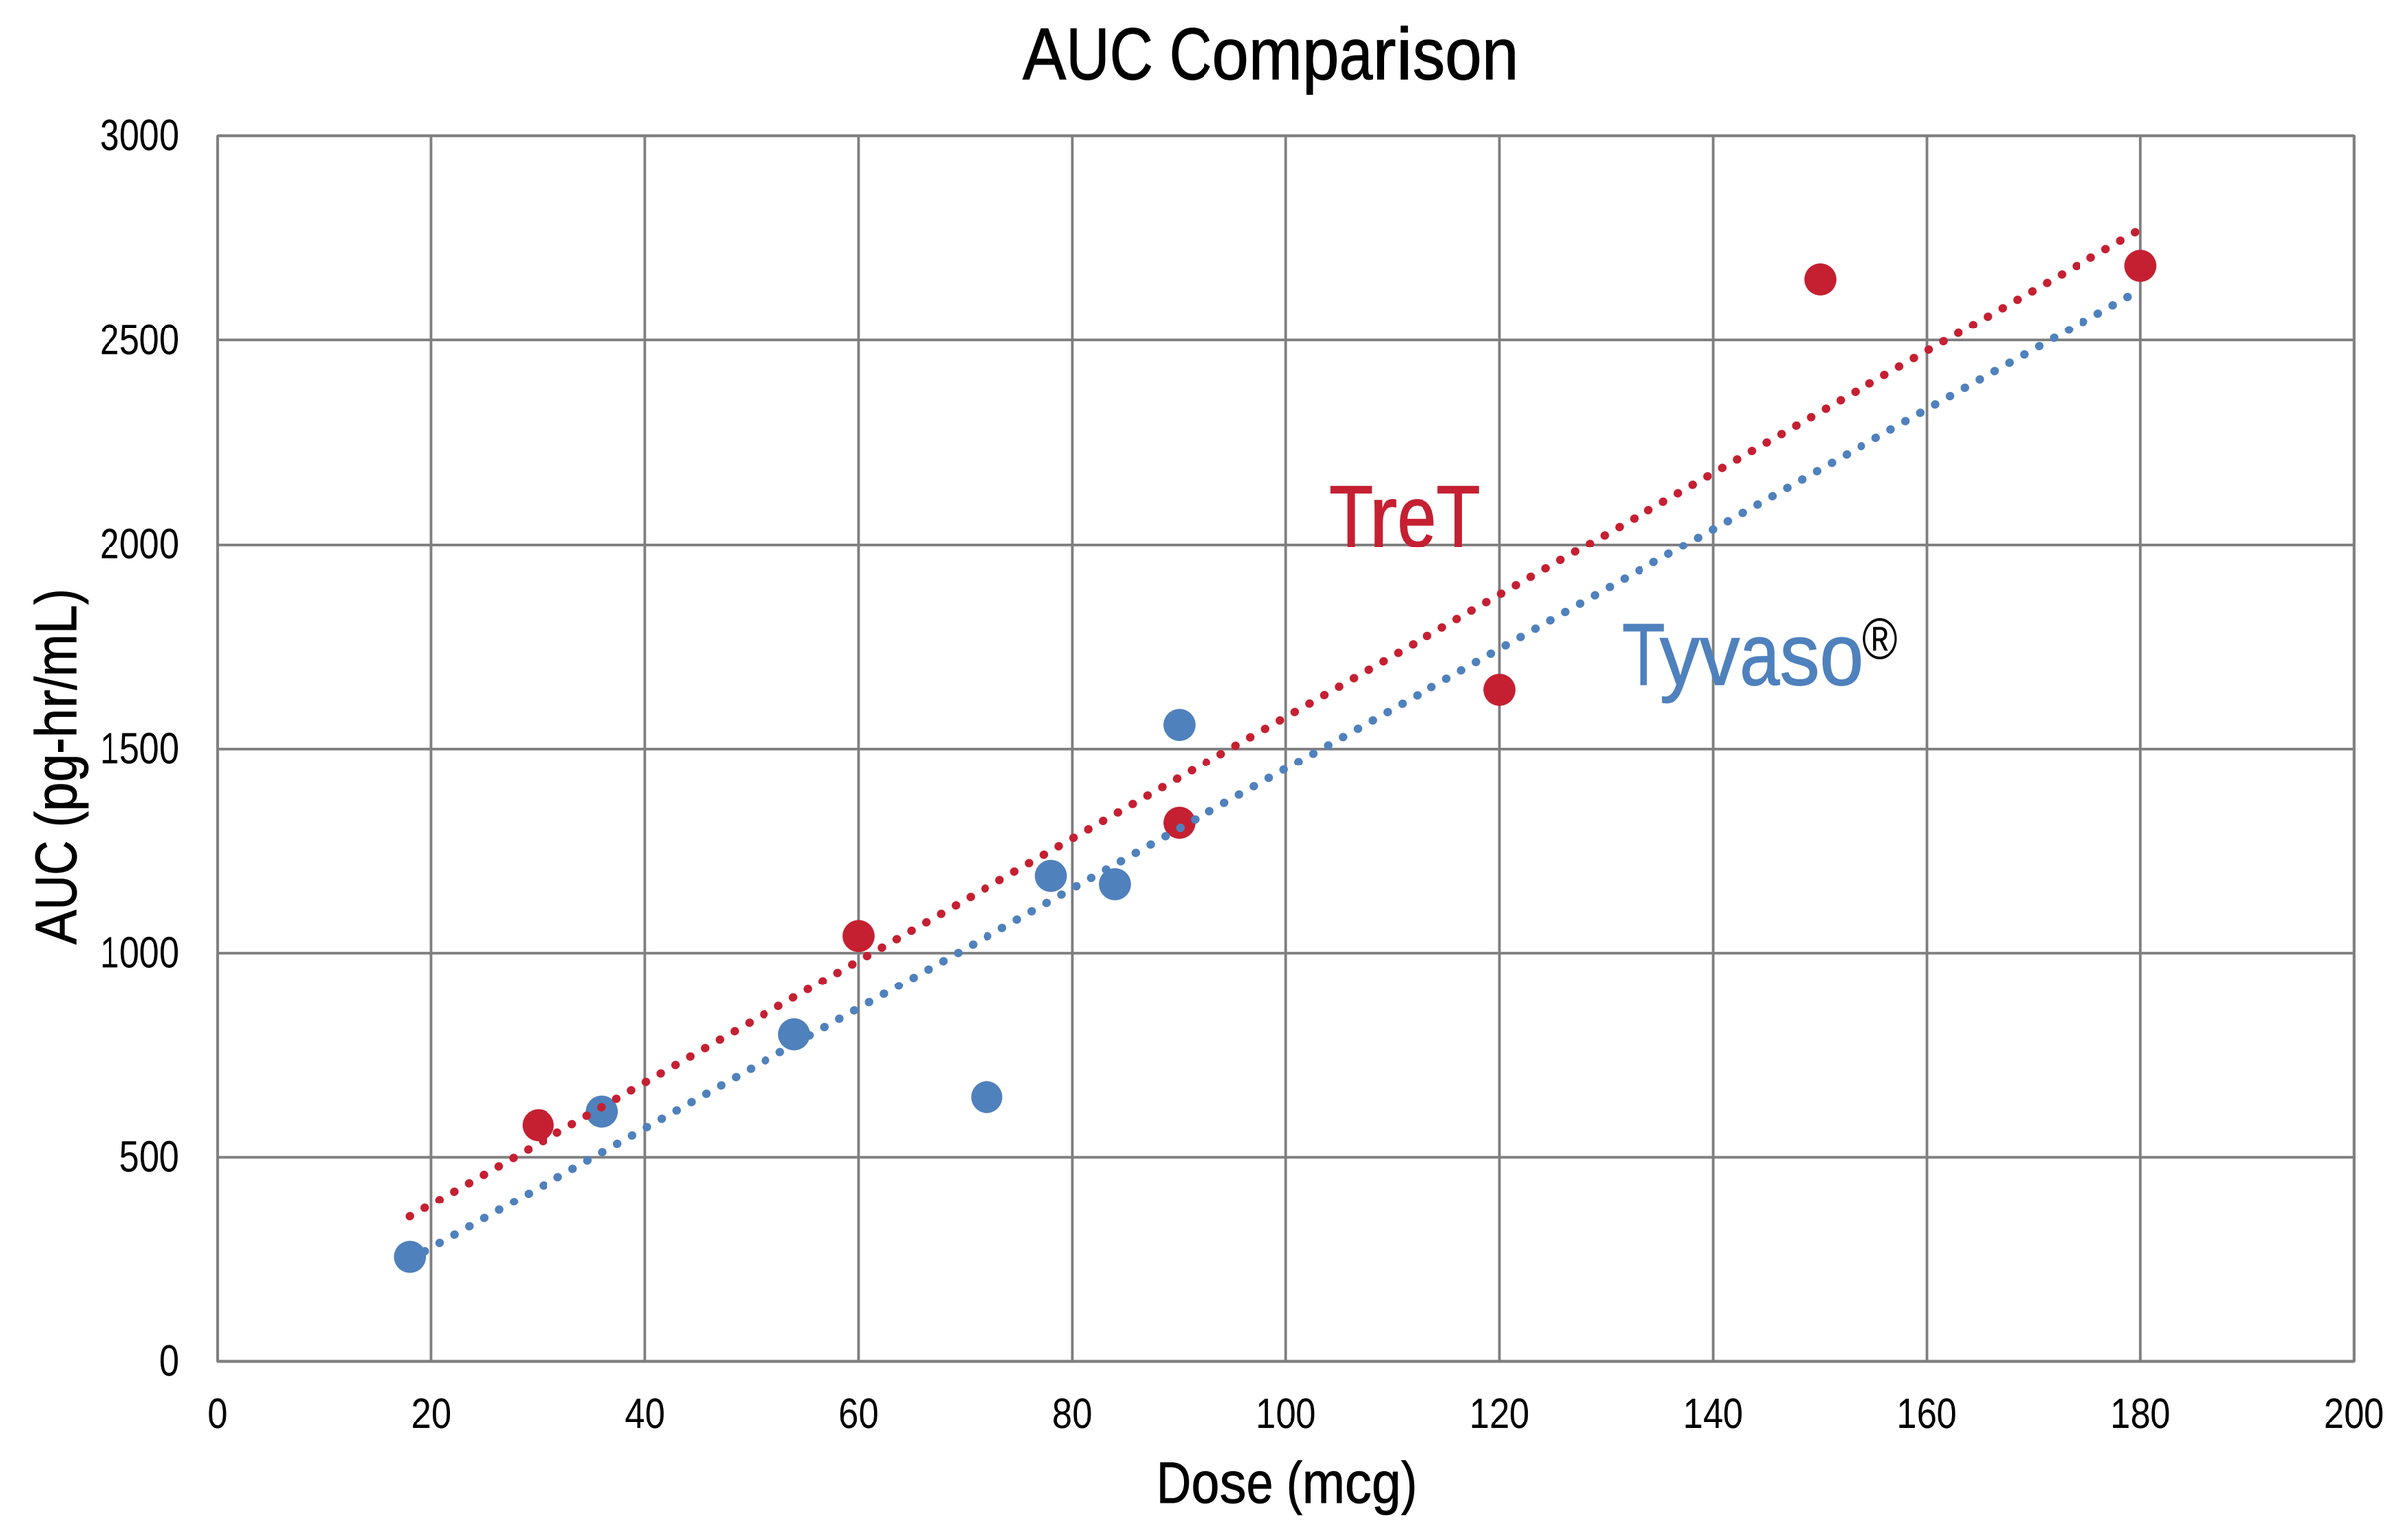
AUC Comparison
AUC (pg-hr/mL)
0
500
1000
1500
2000
2500
3000
Dose (mcg)
0
20
40
60
80
100
120
140
160
180
200
TreT
Tyvaso®

LIMITATIONS

- This study was conducted using single doses in healthy volunteers, further characterization will be done with multiple doses in patients with PAH.

CONCLUSIONS

- Overall, TreT was safe and well-tolerated and produced clinically relevant concentrations of treprostinil when inhaled as a dry powder.
- TreT is currently being evaluated in BREEZE (NCT03950739), a safety and tolerability study, in which 45 patients with PAH on a stable regimen of Tyvaso® will switch to an equivalent dose of TreT. Enrollment in the BREEZE study is ongoing.

REFERENCES

1. Simonneau G, Gatzoulis MA, Adatia I, et al. Updated clinical classification of pulmonary hypertension. *J Am Coll Cardiol*. 2013;62(25 Suppl):D34-D41.
2. Tyvaso: RIN-PH-102 and TRIUMPH BIO data on file.
3. TreT: Table 14.4.2 PK Analysis Set provided by MannKind data on file.

Presented at European Respiratory Society International Conference | September 27-October 2, 2019 | Madrid, Spain | #PA4749
Disclosure: This study was sponsored by United Therapeutics Corporation.

United Therapeutics CORPORATION