Clinical Improvement in Patients Treated with Oral Treprostinil: A Post Hoc Analysis of the FREEDOM-EV Study

Sandeep Sahay, MD[1]; Franck F. Rahaghi, MD[2]; Karim El-Kersh, MD[3]; Akram Khan, MD[4]; Margaret R. Sketch, PharmD, MPH[5]; Meredith Broderick, PharmD, JD[5]; Louis Holdstock, PhD[5]; Youlan Rao, PhD[5]; Eunah Cho, BA[5]; Raymond Benza, MD, FACC[6]

[1]Weill Cornell Medical College, Institute of Academic Medicine, Houston Methodist Lung Center, Houston Methodist Hospital, Houston, TX; [2]Cleveland Clinic Florida, Weston, FL; [3]University of Louisville School of Medicine, Louisville, KY; [4]Oregon Health and Science University, Portland, OR; [5]United Therapeutics Corporation, Research Triangle Park, NC; [6]The Ohio State University Wexner Medical Center, Columbus, OH

BACKGROUND

- Oral treprostinil is an oral prostacyclin analogue indicated to improve exercise capacity and delay disease progression in patients with pulmonary arterial hypertension (PAH)
- Contemporary clinical trials in PAH have utilized clinical worsening as a primary endpoint
- The FREEDOM-EV study was an international, multicenter, randomized, double-blind, placebo-controlled study that demonstrated addition of oral treprostinil reduced the risk of clinical worsening by 25% in patients on background monotherapy[1]
- Recent treatment guidelines in PAH have established treatment goals focusing on improvement to or maintenance of low risk profile according to relevant clinical parameters, emphasizing the importance of improvement over stability[2]
- Clinical improvement as a composite endpoint has been put forth in the REPLACE study, and has been applied post hoc to other clinical trials in PAH[3,4]

AIM

- This post hoc analysis assessed clinical improvement, as defined in the REPLACE study, as a composite endpoint in patients with PAH in the FREEDOM-EV study.

METHODS

- In FREEDOM-EV, patients received were randomized to receive placebo (n=344) or oral treprostinil (n=346) titrated up from 0.125 mg three times daily (TID) to a maximum dose of 12 mg TID. All patients were on background oral monotherapy at study entry.
- The proportion of patients achieving clinical improvement was assessed at Week 24, 36, and 60 in the FREEDOM-EV study
- Per the REPLACE study, clinical improvement was defined as the absence of clinical worsening and at least 2 of the 3:
 - ≥10% or ≥30 m improvement in 6MWD from baseline
 - Improvement to or maintenance of WHO functional class (FC) I/II
 - ≥30% reduction in NT-proBNP from baseline
- Clinical worsening was defined as death, hospitalization due to worsening PAH, initiation of an inhaled or infused prostacyclin for the treatment of worsening PAH, disease progression, or unsatisfactory long-term clinical response
- Missing data were imputed using worst value or last observation carried forward (LOCF) approach, as appropriate. No imputation was performed at Weeks 36 and 60.
- Odds ratio (OR) and p-value from logistic regression adjusted by baseline French noninvasive low-risk criteria. The baseline French noninvasive low-risk is included in the logistic regression model as a covariate to adjust for the imbalance in risk between the treatment and placebo arms at baseline.
- In the forest plots, subgroup is additionally considered in the logistic regression for each subgroup. A Wald statistic is used to construct the 95% confidence interval.

RESULTS

Figure 1. Proportion of patients achieving clinical improvement at Weeks 24, 36, and 60 in FREEDOM-EV

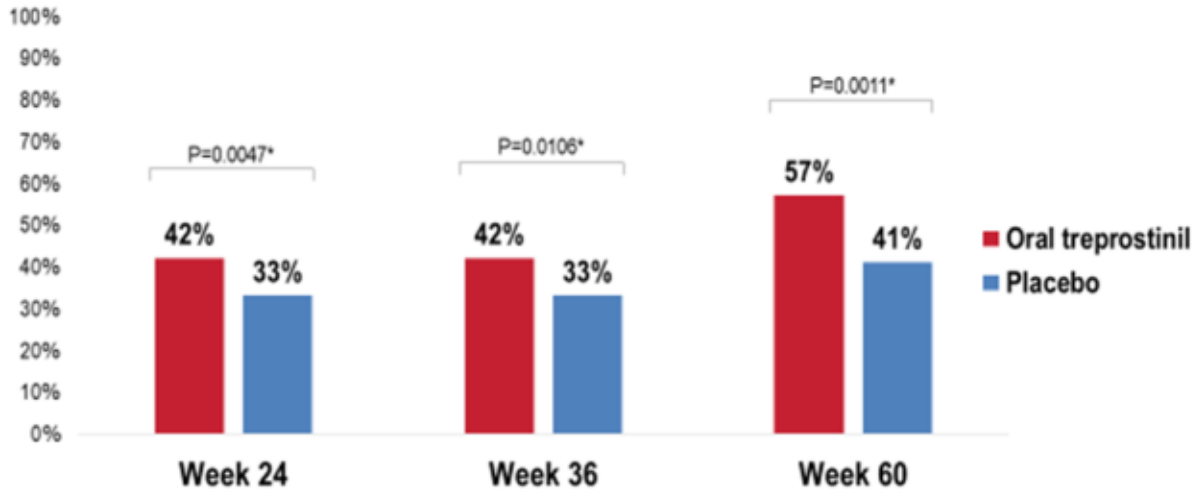
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
P=0.0047*
P=0.0106*
P=0.0011*
42%
33%
42%
33%
57%
41%
Week 24
Week 36
Week 60
Oral treprostinil
Placebo

* P-value by CMH test stratified by baseline noninvasive French low-risk criteria.

Table 1. Clinical improvement at Weeks 24, 36, and 60 in FREEDOM-EV

	Oral Treprostinil	Placebo	P-Value‡
Week 24	N=346	N=344	
Clinical Improvement – n (%)	144 (42)	115 (33)	0.0047*
Odds ratio (95% CI)‡	1.594 (1.154, 2.202)		0.0047
Week 36	N=328	N=329	
Clinical Improvement – n (%)	138 (42)	110 (33)	0.0106*
Odds ratio (95% CI)‡	1.537 (1.106, 2.137)		0.0105
Week 60	N=184	N=189	
Clinical Improvement – n (%)	104 (57)	77 (41)	0.0011*
Odds ratio (95% CI)‡	2.023 (1.325, 3.087)		0.0011

* P-value by CMH test stratified by baseline noninvasive French low-risk criteria. ‡ OR (95% CI) and P-value from logistic regression adjusted by noninvasive French low-risk criteria.

Table 2. Proportion of patients achieving individual components of clinical improvement at Weeks 24, 36, and 60

	Week 24		Week 36		Week 60	
	TRE (N=346)	PBO (N=344)	TRE (N=328)	PBO (N=329)	TRE (N=184)	PBO (N=189)
No clinical worsening	95%	88%	90%	82%	98%	97%
6MWD increase by ≥10% or ≥30 m from baseline	33%	30%	37%	30%	45%	36%
WHO FC I/II	66%	67%	64%	62%	80%	78%
NT-proBNP reduction of ≥30% from baseline	36%	22%	34%	22%	43%	23%

TRE = oral treprostinil, PBO = placebo

Figure 2. Forest plot on subgroup analyses at Week 24

Subgroup	Oral treprostinil Clin. Improv.	Oral treprostinil Total	Placebo Clin. Improv.	Placebo Total	OR	95% CI Lower	95% CI Upper
Overall	144	346	115	344	1.594	1.154	2.202
Age Group							
< 43 years (median age)	80	166	62	173	1.700	1.090	2.651
≥ 43 years (median age)	64	180	53	171	1.609	0.993	2.608
Baseline 6MWD							
≤ 350 m	31	95	36	93	0.877	0.467	1.647
> 350 m	113	251	79	251	2.044	1.394	2.997
Baseline WHO FC							
WHO FC I or II	107	214	93	241	1.631	1.113	2.389
WHO FC III or IV	37	132	22	103	1.491	0.799	2.782
PAH Etiology							
Idiopathic or Heritable	104	219	74	216	1.981	1.316	2.982
Collagen Vascular Disease	29	94	24	84	1.166	0.603	2.257
Other	11	33	17	44	1.336	0.441	4.048
Background PAH Therapy							
PDE-5I or sGC Stimulator	107	248	85	246	1.603	1.095	2.346
ERA	37	98	30	98	1.764	0.932	3.338
Dose at 24 weeks							
<3 mg tid	45	106	13	35	1.631	0.704	3.775
≥ 3 mg tid	93	182	100	254	1.878	1.253	2.815
Baseline NT-proBNP							
< 300 pg/mL	51	126	62	163	1.237	0.749	2.044
≥ 300 pg/mL	93	220	53	181	1.893	1.235	2.902
Time Since PAH Diagnosis							
< 6 months	76	170	64	163	1.421	0.903	2.236
≥ 6 months	68	176	51	181	1.821	1.141	2.908
Duration on PAH Background Therapy							
< 6 months	91	193	67	179	1.643	1.070	2.522
≥ 6 months	53	153	48	165	1.526	0.925	2.519

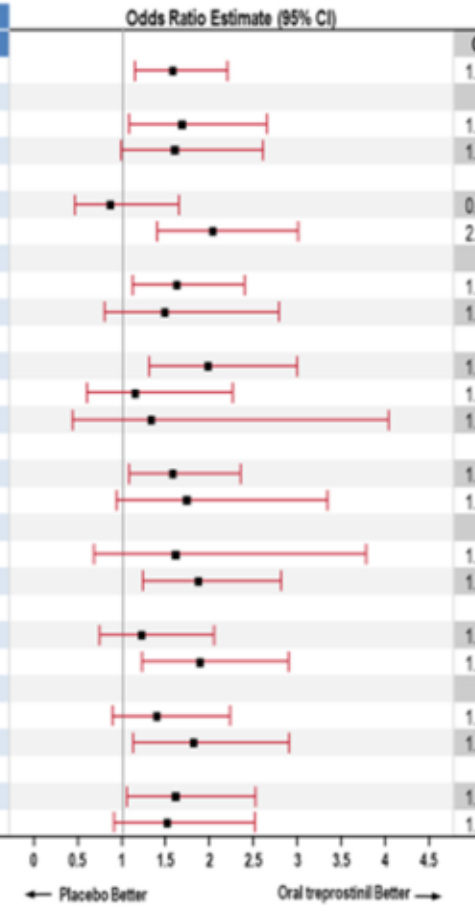
Odds Ratio Estimate (95% CI)
0
0.5
1
1.5
2
2.5
3
3.5
4
4.5
← Placebo Better
Oral treprostinil Better →

Figure 3. Forest plot on subgroup analyses at Week 36

Subgroup	Oral treprostinil Clin. Improv.	Oral treprostinil Total	Placebo Clin. Improv.	Placebo Total	OR	95% CI Lower	95% CI Upper
Overall	138	328	110	329	1.537	1.106	2.137
Age Group							
< 43 years (median age)	79	158	60	167	1.803	1.145	2.839
≥ 43 years (median age)	59	170	50	162	1.330	0.817	2.163
Baseline 6MWD							
≤ 350 m	29	91	34	91	0.905	0.473	1.731
> 350 m	109	237	76	238	1.906	1.293	2.809
Baseline WHO FC							
WHO FC I or II	100	203	87	230	1.583	1.069	2.342
WHO FC III or IV	38	125	23	99	1.438	0.776	2.663
PAH Etiology							
Idiopathic or Heritable	100	213	65	205	2.115	1.387	3.224
Collagen Vascular Disease	29	89	27	82	1.035	0.534	2.003
Other	9	26	18	42	0.812	0.267	2.475
Background PAH Therapy							
PDE-5I or sGC Stimulator	103	235	84	236	1.505	1.019	2.223
ERA	35	93	26	93	1.855	0.968	3.556
Dose at 24 weeks							
<3 mg tid	32	73	7	13	0.849	0.237	3.043
≥ 3 mg tid	92	156	95	225	2.104	1.365	3.244
Baseline NT-proBNP							
< 300 pg/mL	52	118	62	154	1.155	0.694	1.925
≥ 300 pg/mL	86	210	48	175	1.916	1.237	2.968
Time Since PAH Diagnosis							
< 6 months	75	161	60	156	1.526	0.958	2.432
≥ 6 months	63	167	50	173	1.630	1.012	2.626
Duration on PAH Background Therapy							
< 6 months	86	181	61	169	1.676	1.074	2.615
≥ 6 months	52	147	49	160	1.380	0.837	2.275

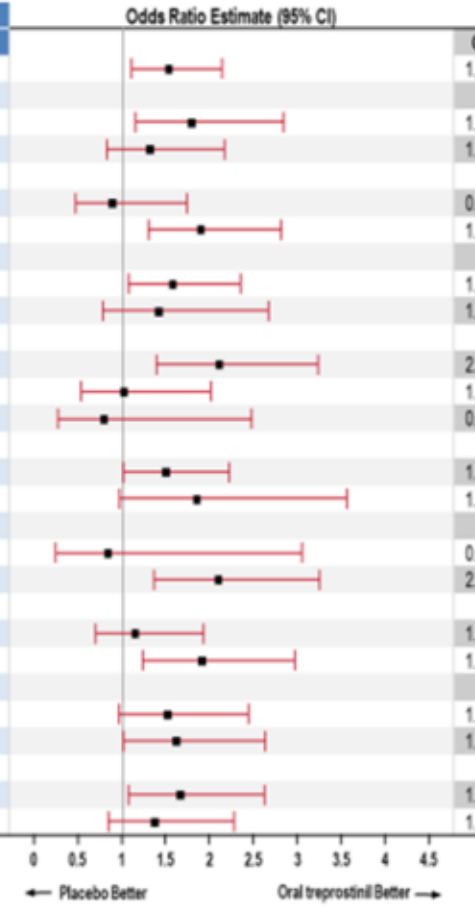
Odds Ratio Estimate (95% CI)
0
0.5
1
1.5
2
2.5
3
3.5
4
4.5
← Placebo Better
Oral treprostinil Better →

RESULTS (cont.)

- At both Weeks 24 and 36, 42% of patients in the oral treprostinil group achieved clinical improvement compared to 33% in the placebo group, and at Week 60, 57% of patients achieved clinical improvement in the oral treprostinil group compared to 41% in the placebo group, when adjusting by baseline French noninvasive low-risk criteria
- Patients treated with oral treprostinil were 1.59, 1.54, and 2.02 times more likely to achieve clinical improvement at Weeks 24, 36, and 60 compared to the placebo group, respectively, when adjusting by French noninvasive low risk criteria
- Individual components the clinical improvement definition
 - >90% of patients in the oral treprostinil group had no clinical worsening at all time points evaluated
 - A majority of patients in the placebo and oral treprostinil groups were WHO FC I/II at each time point
 - The largest difference in individual components was the proportion of patients achieving ≥30% reduction in NT-proBNP from baseline in the oral treprostinil group compared to the placebo group
- Clinical improvement was generally consistent across patient subgroups at Weeks 24, 36, and 60 [not shown]. Patient characteristics in the oral treprostinil group favoring likelihood of achieving clinical improvement were younger age, idiopathic/heritable PAH, PDE-5i/sGC stimulator background therapy, baseline 6MWD >350 m, baseline WHO FC I/II, baseline NT-proBNP ≥300 pg/mL, dose ≥3 mg TID at Week 24, or <6 months on background PAH therapy.

CONCLUSIONS

- The addition of oral treprostinil on top of background monotherapy increased the proportion and likelihood of patients achieving clinical improvement, as defined by the REPLACE study, at Weeks 24, 36, and 60 in the FREEDOM-EV study.
- Oral treprostinil had the most robust effect on NT-proBNP compared to placebo.
- Notable subgroups of the oral treprostinil group that had a more favorable response when assessing clinical improvement at all time points were patients achieving a dose of ≥3 mg TID by 24 weeks or those on PDE5-i/sGC stimulator background therapy.
- These findings suggest that in addition to delaying clinical worsening, treatment with oral treprostinil on background monotherapy improved clinical status in patients as early as 24 weeks, with maintained effect through 60 weeks.
- Further research is needed to understand application of clinical improvement in real-world practice and explore its association with long-term outcomes.

REFERENCES

1. White RJ, *et al.* Combination Therapy with Oral Treprostinil for Pulmonary Arterial Hypertension. A Double-Blind Placebo-controlled Clinical Trial. *Am J Respir Crit Care Med.* 2020;201(6):707-17.
2. Galie N, *et al.* 2015 ESC/ERS Guidelines for the diagnosis and treatment of pulmonary hypertension: hypertension: The Joint Task Force for the Diagnosis and Treatment of Pulmonary Hypertension of the European Society of Cardiology (ESC) and the European Respiratory Society (ERS). *Eur Heart J.* 2016; 37: 67–119.
3. Hoeper MM, *et al.* witching to riociguat versus maintenance therapy with phosphodiesterase-5 inhibitors in patients with pulmonary arterial hypertension (REPLACE): a multicentre, open-label, randomised controlled trial. *Lancet Respir Med.* 2021; ePub ahead of print.
4. Simonneau G, *et al.* Assessment of the REPLACE study composite endpoint in riociguat-treated patients in the PATENT study. *Pulmonary Circulation.* 2020; 10(4).

Presented at the ATS International Conference, May 14-19, 2021 Virtual | poster A3601
Disclosures: This study was sponsored by United Therapeutics Corporation.

United Therapeutics CORPORATION