Interim Results from the Ongoing Open-Label Extension of the Phase 2 Trial Evaluating Ralinepag for the Treatment of Pulmonary Arterial Hypertension

V. McLaughlin, MD[1]; A. Ristic, MD, PhD[2]; P. Jansa, MUDr, PhD[3]; E.S. Klings, MD[4]; J. Barberá, MD, PhD[5]; A. Keogh, MD[6]; J. Adams, PhD[7]; B. Walsh, PhD[7]; J. Zhang[7]; C. Cabell, MD[7]; P. Klassen, MD[7]; L. Rubin, MD[8,9]

[1]University of Michigan, Ann Arbor, MI; [2]Belgrade University School of Medicine, Belgrade, Serbia; [3]Charles University, Prague, Czech Republic; [4]Boston University School of Medicine, Boston, MA; [5]University of Barcelona, Barcelona, Spain; [6]St. Vincent's Hospital, Sydney, Australia; [7]Arena Pharmaceuticals, San Diego, CA; [8]University of California, La Jolla, CA; [9]Columbia University College of Physicians and Surgeons, New York, NY

INTRODUCTION

- Ralinepag is a novel, next-generation, oral, selective, potent prostacyclin (IP) receptor agonist in development for pulmonary arterial hypertension (PAH), with optimized pharmacokinetics (PK) and potent activity on pulmonary arteries, vascular smooth muscle cells, and platelets.
- APD811-003 was a Phase 2 randomized, double-blind, placebo-controlled clinical study that assessed the effects of ralinepag on hemodynamics and on 6-minute walk distance (6MWD) in 61 subjects with PAH after 22-weeks of treatment.
- In APD811-003, the median change in pulmonary vascular resistance (PVR) with ralinepag was –163.9 dyn•s•cm^{-5} (baseline, 705 dyn•s•cm^{-5}) compared with an increase of 0.7 dyn•s•cm^{-5} (baseline, 480 dyn•s•cm^{-5}) in patients receiving placebo (p=0.02). These improvements were observed in patients receiving PAH-specific background therapy (59% dual; 41% mono).
- Subjects who completed APD811-003 were able to enroll in APD811-007, a multicenter, open-label extension (OLE) study evaluating the long-term safety and tolerability of ralinepag in patients with PAH who completed APD811-003 or who were assigned to placebo and were discontinued for clinical worsening.

METHODS

Figure 1. APD811-003 and APD811-007 Study Design

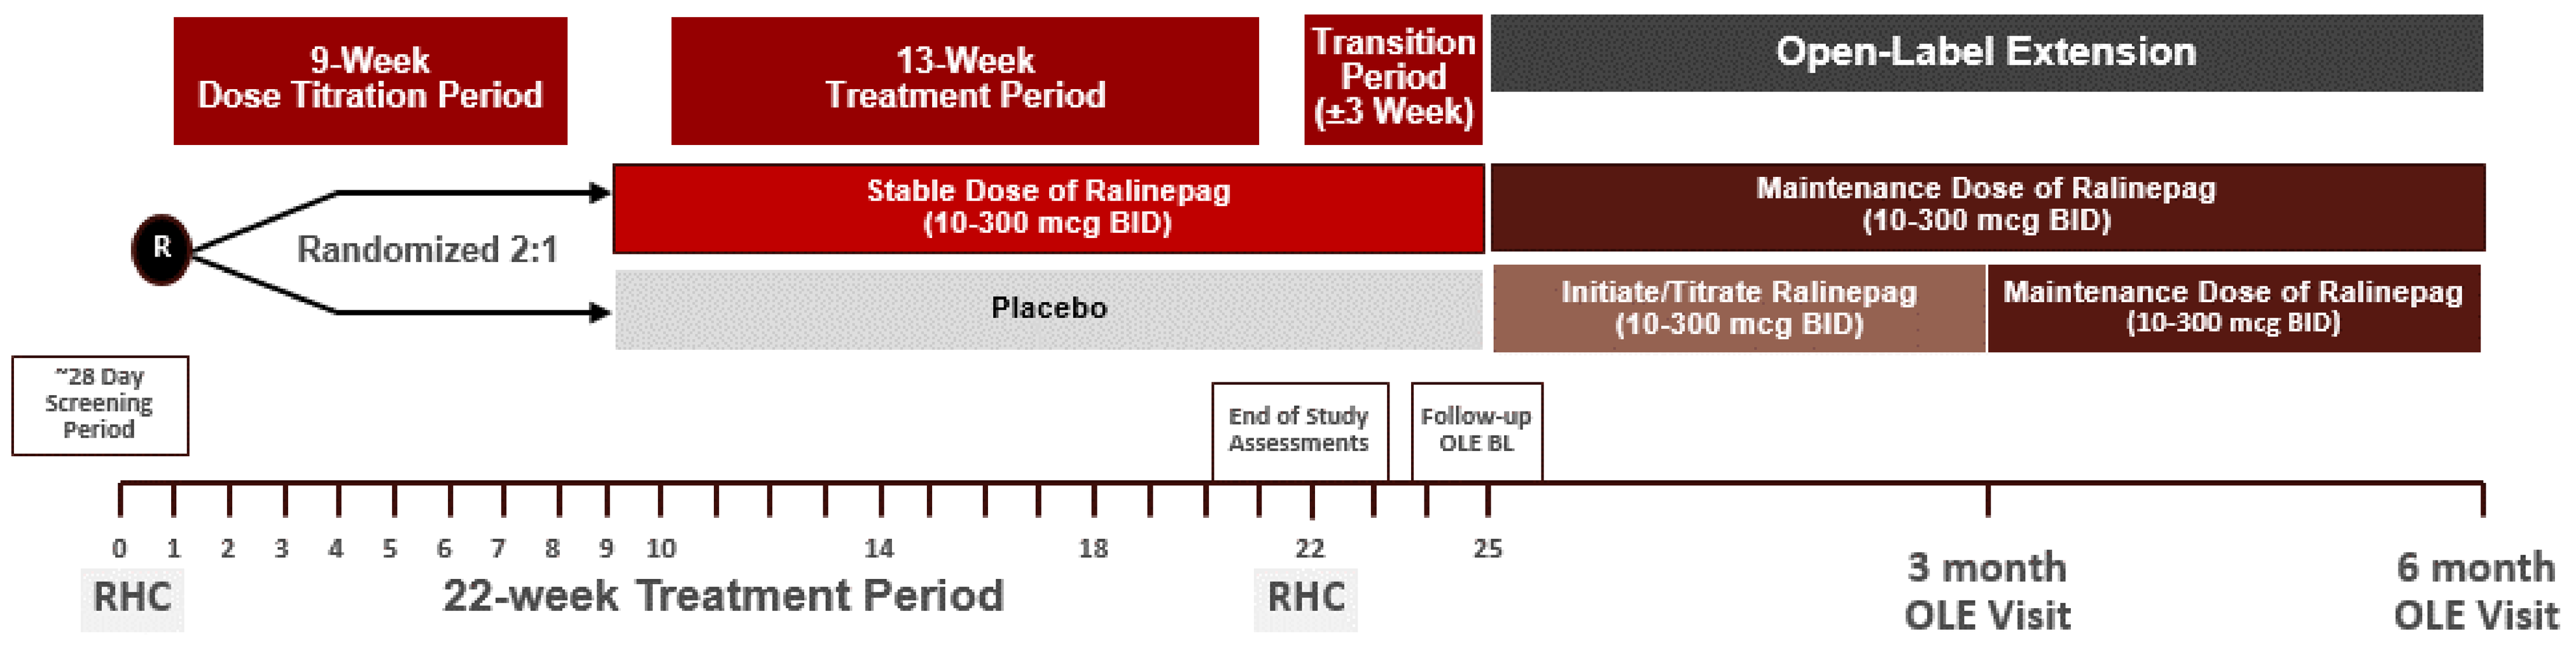
9-Week
Dose Titration Period
13-Week
Treatment Period
Transition
Period
(±3 Week)
Open-Label Extension
R
Randomized 2:1
Stable Dose of Ralinepag
(10-300 mcg BID)
Maintenance Dose of Ralinepag
(10-300 mcg BID)
Placebo
Initiate/Titrate Ralinepag
(10-300 mcg BID)
Maintenance Dose of Ralinepag
(10-300 mcg BID)
~28 Day
Screening
Period
End of Study
Assessments
Follow-up
OLE BL
0 1 2 3 4 5 6 7 8 9 10 14 18 22 25
RHC
22-week Treatment Period
RHC
3 month
OLE Visit
6 month
OLE Visit

- Of the 61 subjects in the Phase 2 study, 45 continued into the open-label extension study. Subjects randomized to ralinepag continued on active therapy (n=30), and subjects originally randomized to placebo switched to ralinepag (n=15) and titrated drug weekly for 9-weeks until a stable maximum tolerated dose was reached.
- Subjects visited the clinic monthly for the first 3 months, then every 3 months until subject discontinuation or study termination.
- Key safety assessments included clinical laboratory tests, vital signs, physical examinations, 12-lead electrocardiograms, and adverse events (AEs). Key efficacy assessments included PVR and 6MWD.
- All data was analyzed using the signed rank test.

BASELINE DEMOGRAPHICS

	Ralinepag/Ralinepag (n=30)	Placebo/Ralinepag (n=15)	All Patients (n=45)
Age (years), median (range)	46 (19, 68)	60 (34, 71)	51 (19, 71)
Female/Male (%)	80/20	100/0	87/13
PAH Duration, median (range)	2.5 (0.3-27)	1.8 (0.3-14)	2.0 (0.3-27)
PAH Concomitant Medication, n (%)			
PDE5 Inhibitor Monotherapy	10 (33%)	4 (27%)	14 (31%)
ERA Monotherapy	1 (3%)	4 (27%)	5 (11%)
ERA + PDE5 Inhibitor/sGC	19 (63%)	7 (47%)	26 (58%)
PVR (dyn•s•cm^{-5}), median (range)	732 (343, 2119)	480 (282, 1050)	576 (282, 2119)
6MWD (m), median (range)	422 (105, 686)	367 (180, 543)	415 (105, 686)

RESULTS

- Of the 30 subjects who continued ralinepag in the OLE, 18 underwent right heart catheterization (RHC) (median treatment duration at RHC, 1.8 years; range 1.2-3.4 years). In the subjects originally on ralinepag, improvements in the original study were sustained for PVR (N=18; median reduction from APD811-003 baseline, 219 dyn•s•cm^{-5} [range, -779 to 534 dyn•s•cm^{-5}]; APD811-003 baseline PVR, 515 dyn•s•cm^{-5}, [range, -779 to 534 dyn•s•cm^{-5}]; p=0.002). Compared to APD811-007 baseline, median reduction was 46.5 dyn•s•cm^{-5} (range, -370 to 415 dyn•s•cm^{-5}).
- In patients who switched from placebo to ralinepag, 11/15 underwent RHC (median treatment duration at RHC, 1.4 years; range, 0.9–2.3 years). Similar magnitudes of improvement were observed from baseline in the original study for PVR (median reduction, 214 dyn•s•cm^{-5} [range, -404 to 552 dyn•s•cm^{-5}]; baseline PVR, 467 dyn•s•cm^{-5}; p=0.206). Compared to APD811-007 baseline, median reduction was 103 dyn•s•cm^{-5} (range, -573 to 230 dyn•s•cm^{-5}).

Figure 2. Change in PVR in APD811-003 and APD811-007

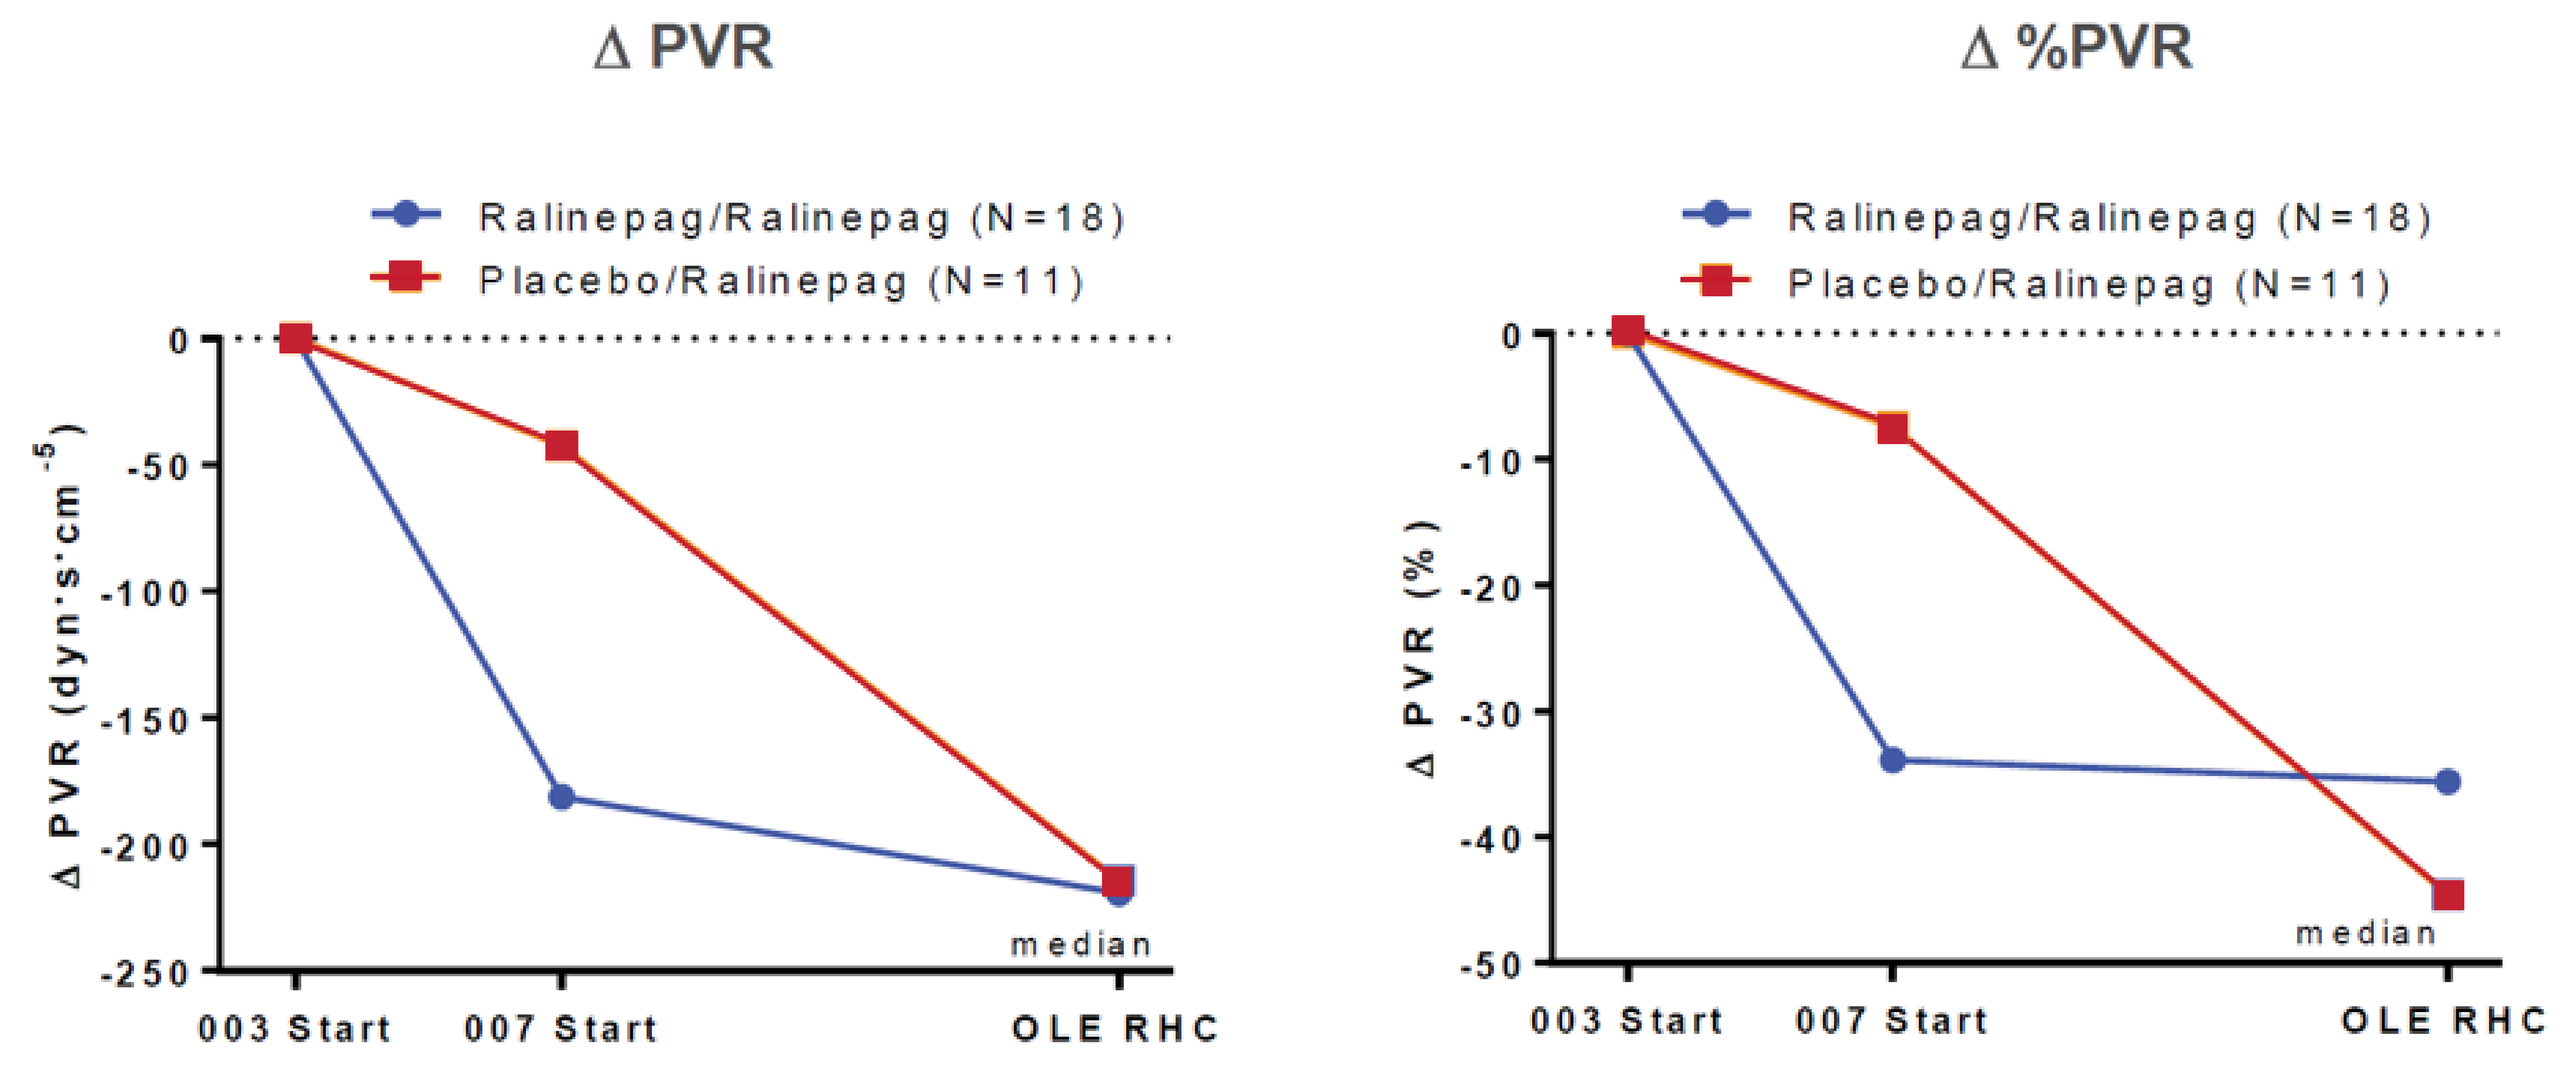
Δ PVR
Ralinepag/Ralinepag (N=18)
Placebo/Ralinepag (N=11)
Δ PVR (dyn·s·cm-5)
0
-50
-100
-150
-200
-250
median
003 Start
007 Start
OLE RHC
Δ %PVR
Ralinepag/Ralinepag (N=18)
Placebo/Ralinepag (N=11)
Δ PVR (%)
0
-10
-20
-30
-40
-50
median
003 Start
007 Start
OLE RHC

RESULTS

Figure 3. Change in 6MWD Over Time

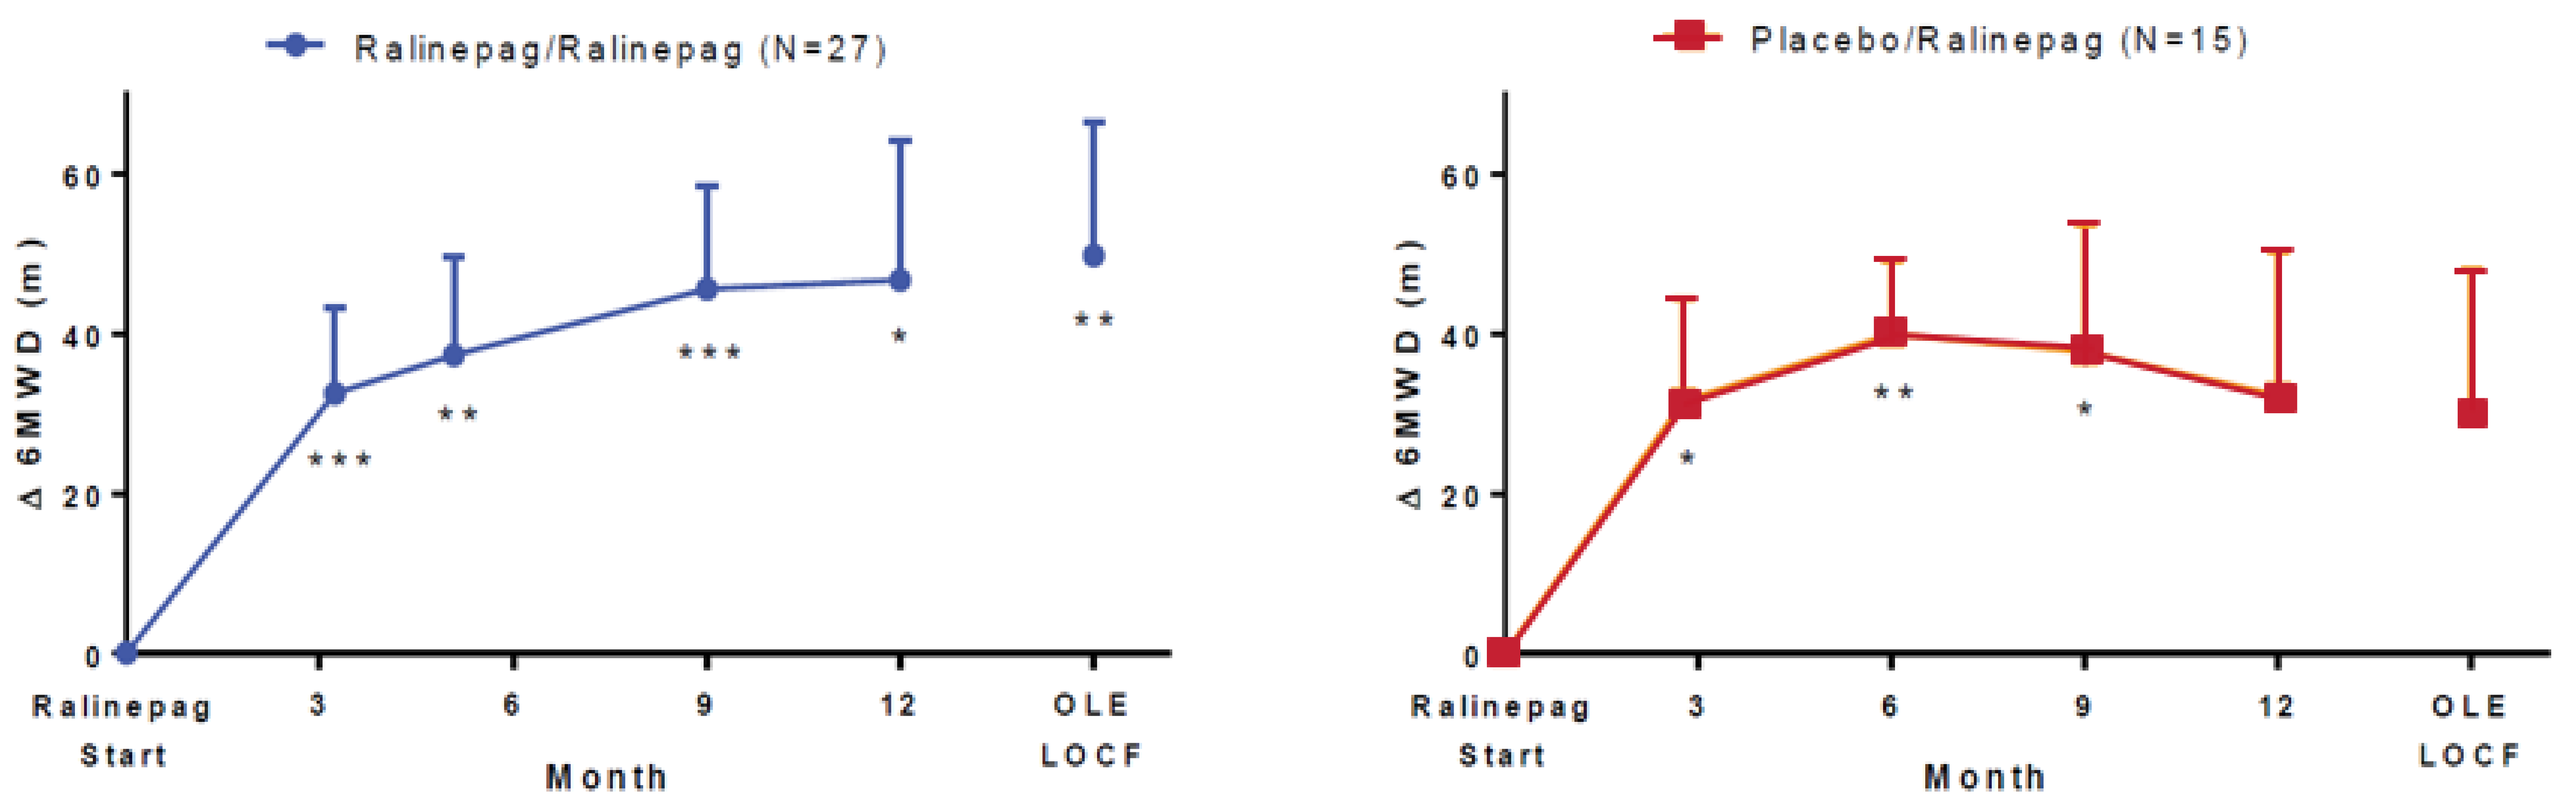
Ralinepag/Ralinepag (N=27)
Δ 6MWD (m)
60
40
20
0

**

*
**
Ralinepag
Start
3
6
9
12
OLE
LOCF
Month
Placebo/Ralinepag (N=15)
Δ 6MWD (m)
60
40
20
0
*
**
*
Ralinepag
Start
3
6
9
12
OLE
LOCF
Month

- In the subjects who continued ralinepag in the OLE, median improvement in 6MWD (last observation) from APD811-003 baseline was 31 meters (n=27; range: -2 to 62 meters; baseline 425 meters; p=0.003) and from APD811-007 baseline was 12 meters (n=27; range: -80 to 110; baseline 445 meters; p=0.160).
- In the subjects who switched from placebo to ralinepag in the OLE, median improvement in 6MWD (last observation) from APD811-003 baseline was 74.2 meters (n=15; range -168 to 274 meters; baseline 367 meters; p=0.010) and from APD811-007 baseline was 48 meters (n=15; range: -126 to 138 meters; baseline 410 meters; p=0.107).
- The most common AEs in subjects originally randomized to ralinepag were lower in the OLE (headache, 37%; nausea, 17%) versus the original study (headache, 78%; nausea, 50%). The most common AEs in subjects originally randomized to placebo were headache (87%), nausea (40%), myalgia (33%), flushing (27%), and diarrhea (27%).

LIMITATIONS

- This was an open-label study that was not designed to analyze efficacy (PVR and 6MWD). As such, all data analysis presented here is exploratory in nature. Only subjects who completed the RHC and 6MWD tests were included in the analyses.
- Not all subjects consented to and underwent the RHC in the open-label extension, and the timing of the RHC between subjects was variable. The RHC was added in a protocol amendment and was to be done 1 year after enrollment in APD811-007. If the subject had been enrolled longer than 1 year, it was to be done after 2 years. If the subject had been enrolled longer than 2 years, it was to be done as soon as possible.

CONCLUSIONS

- In patients continuing ralinepag, durable, long-term improvements in hemodynamics and 6MWD were observed. Patients who switched from placebo also experienced improvements in PVR and 6MWD. The tolerability and AE profiles were consistent with known effects of prostacyclin receptor agonists.

Presented at ATS 2019 International Conference | May 17-22, 2019 | Dallas, TX | #15260
Disclosure: This studies were sponsored by Arena Pharmaceuticals.